TUSC2 gene therapy in KRAS^{G12C} mutant NSCLC overcomes acquired resistance to sotorasib

AACR-NCI-EORTC Molecular Targets Conference; 2024
Poster# 384

Ismail M Meraz[1], Mourad Majidi[1], Lihui Gao[1], Shuhong Wu[1], Renduo Song[1], Meng Feng[1], Chenghui Ren[1], Yi Xu[1], Qi Wang[2], Yuanxin Xi[2], Jing Wang[2], Sung Yun Jung[4], Elizabeth Shpall[3] and Jack A Roth[1]

Thoracic and Cardiovascular Surgery[1], [2]Bioinformatics and Computational Biology, [3]Stem Cell Transplantation, MD Anderson Cancer Center, Houston, TX, [4]Biochemistry, Baylor College of Medicine, Houston, TX

Abstract

Acquired resistance (AR) to sotorasib, the first FDA-approved KRASi, poses a significant challenge in the treatment of KRAS^{G12C} mutant NSCLC. Despite an initial response, up to 40%, patients invariably develop resistance, necessitating alternative therapeutic strategies. The mechanisms of AR include the emergence of additional mutations in the KRAS gene, reactivation of the KRAS pathway, or activation of alternative signaling pathways. TUSC2, a potent tumor suppressor gene, exhibits multifunctional activities including multikinase inhibition, inhibition of growth & proliferation, induction of cell death & activation of both innate and adaptive immune responses. In this study, we demonstrated that TUSC2 gene therapy effectively overcomes sotorasib AR in KRASG12C mutant NSCLC.

Sotorasib-resistant PDX, CDX, & cell lines were generated. AR-resistant PDXs, CDXs & isogeneic cells were generated by prolonged continuous treatment with sotorasib. Both H23AR & H358AR cells showed >100-fold resistance over sensitive counterparts. These resistant cells also showed resistance to adagrasib, 2nd KRASi. WES performed on AR PDXs, CDX, & cells showed no additional KRAS mutation. RNAseq, RPPA, & mass spectrometry data on TC303AR and TC314AR PDXs showed significant sets of differentially regulated RNA and protein in AR vs. parental. A significant upregulation of PI3K-AKT-mTOR signaling was found in AR PDXs. PI3K and AKT expression were also increased in H23AR and H358AR cells. TUSC2 transfection significantly reduced colony formation in both AR cells. Transfection of TUSC2 also markedly increased the apoptosis in AR cells. H23AR xenograft tumors exhibited significantly lower sensitivity to sotorasib than their parental counterparts. However, treatment with quaratusugene ozeplasmid, TUSC2 loaded nanovesicle, alone or in combination with sotorasib was highly effective in controlling H23AR tumor growth. Quaratusugene ozeplasmid alone also exhibited a significantly strong antitumor effect on TC314AR PDXs whereas Sotorasib showed no significant antitumor activity. However, a synergistic antitumor effect was observed when TC314AR PDX tumors were treated with the combination of quaratusugene ozeplasmid and sotorasib. To further evaluate the antitumor immune responses, immune-competent humanized-NSG mice are generated by transplanting fresh human cord blood-derived CD34+ stem cells into sub-lethally irradiated NSG mice (hu-NSG) to mimic the human immune system with robust engraftment of human CD45, CD3 T, CD19 B, and NK cells. The antitumor immune responses of quaratusugene ozeplasmid on TC314AR PDX tumors are currently being evaluated in the humanized mouse model. In conclusion, TUSC2 therapy, alone or in combination with sotorasib-induced apoptosis, inhibited colony formation, and showed significant antitumor efficacy in KRASG12C mutant-acquired resistant xenograft and PDX tumors.

Development of acquired resistant NSCLC PDXs and isogeneic cells to sotorasib

A

No.	PDX-ID	KRAS	TP53	STK11	PIK3CA	KEAP1
1	TC247	G12C		Splicing		A522P
2	TC303	G12C	R175H			Q292*
3	TC314	G12C	R248L			
4	TC453	G12C		Splicing		
5	TC481	G12C		Y36sf		Y525C
6	TC527	G12C	M160N			
7	TC539	G12C				
8	TC551	G12C				
9	TC569	G12C				
10	TC723	G12C				
11	TC807	G12C		Splicing	G122D	

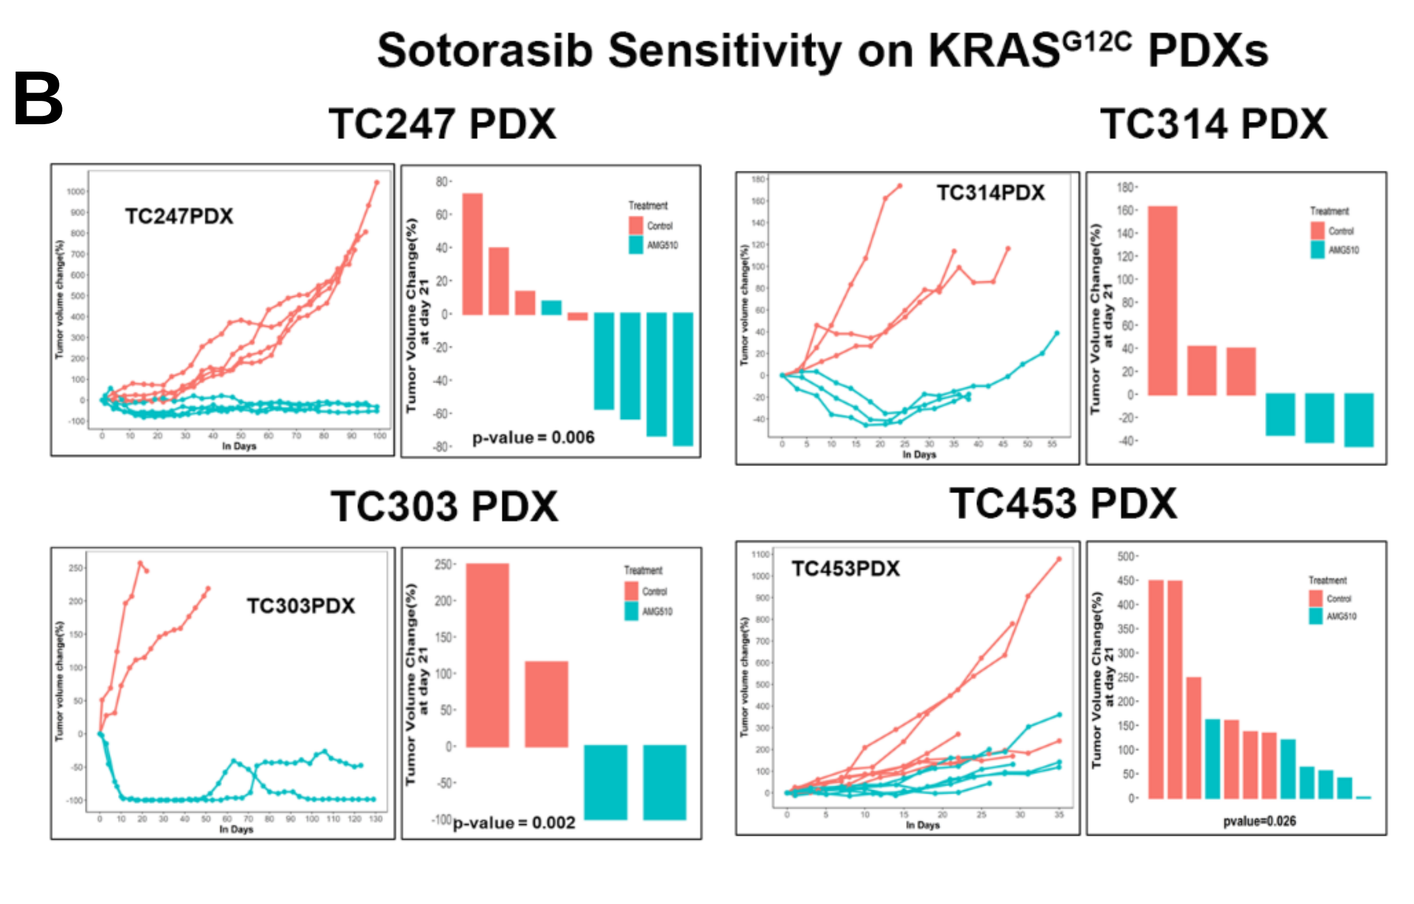
B
Sotorasib Sensitivity on KRAS G12C PDXs
TC247 PDX
TC314 PDX
TC303 PDX
TC453 PDX

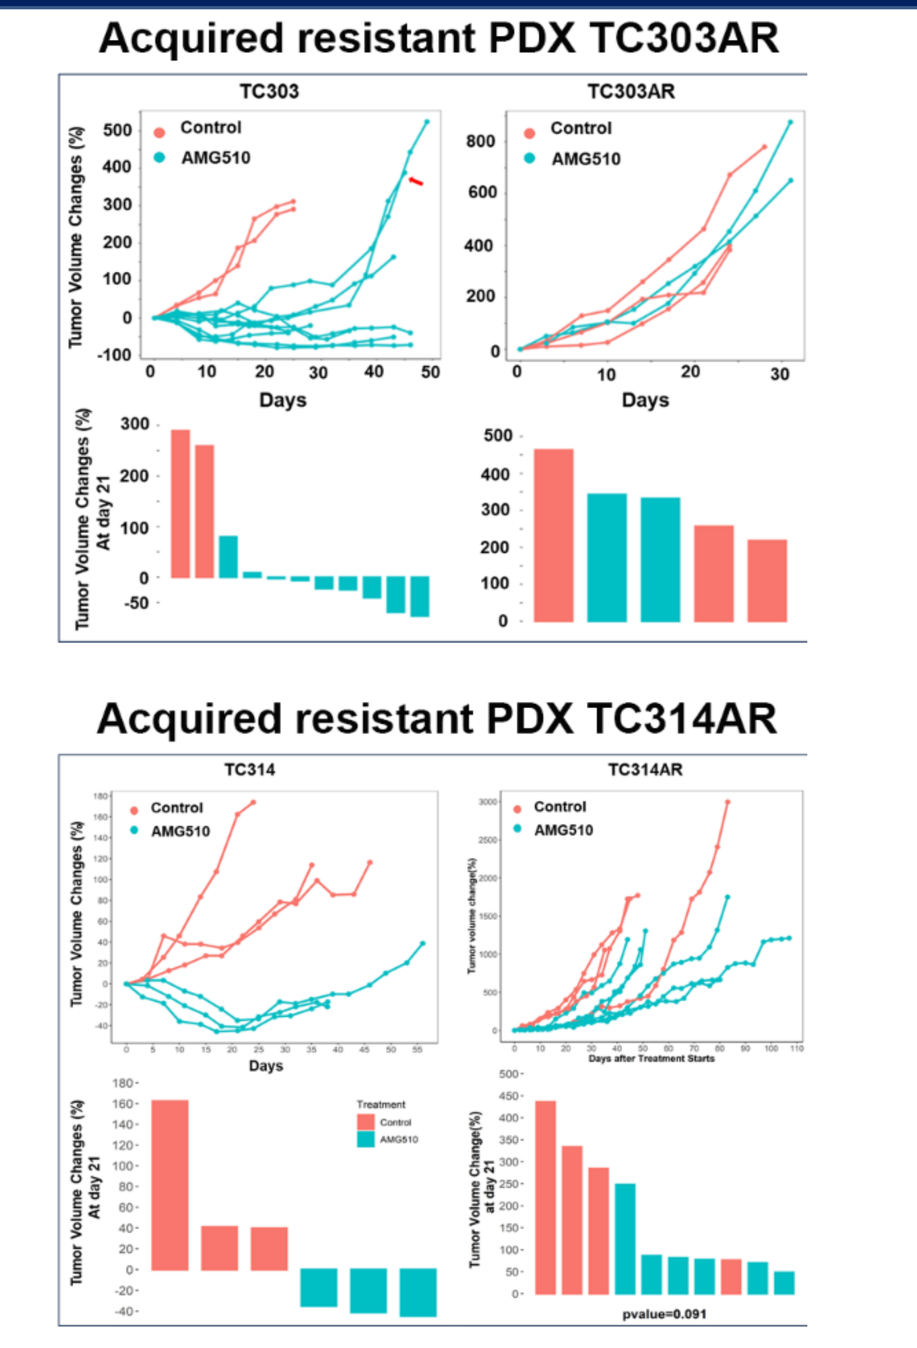
C
Acquired resistant PDX TC303AR
Acquired resistant PDX TC314AR

Fig 1. Development of acquired resistant PDXs. A) The list of KRAS^{G12C} mutant NSCLC PDXs developed in our lab; B) Sensitivity of sotorasib on four KRAS^{G12C} mutant PDXs; C) Development of acquired resistant PDXs TC303AR and TC314AR after prolonged treatment with sotorasib for several generations.

Development of acquired resistant isogeneic NSCLC cell lines

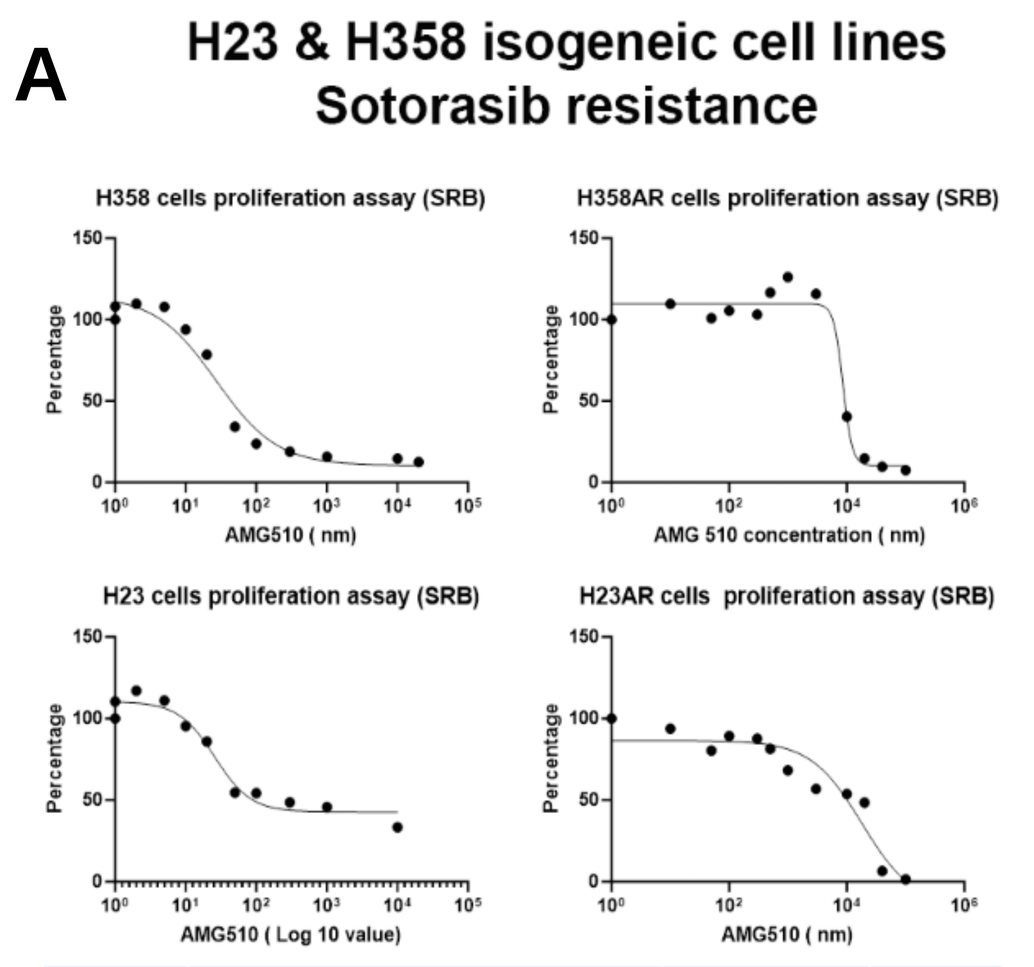
A
H23 & H358 isogeneic cell lines
Sotorasib resistance
H358 cells proliferation assay (SRB)
H358AR cells proliferation assay (SRB)
H23 cells proliferation assay (SRB)
H23AR cells proliferation assay (SRB)

Cell lines	Sotorasib Resistance	IC50 (uM)	Fold Difference
H23	Parental (Sensitive)	0.031 ± 0.007	
H23AR	Isogenic clone (Acquired Resistant)	20.118 ± 1.680	633.8
H358	Parental (Sensitive)	0.027 ± 0.001	
H358AR	Isogeneic cell line (Acquired Resistant)	6.234 ± 1.724	230.5

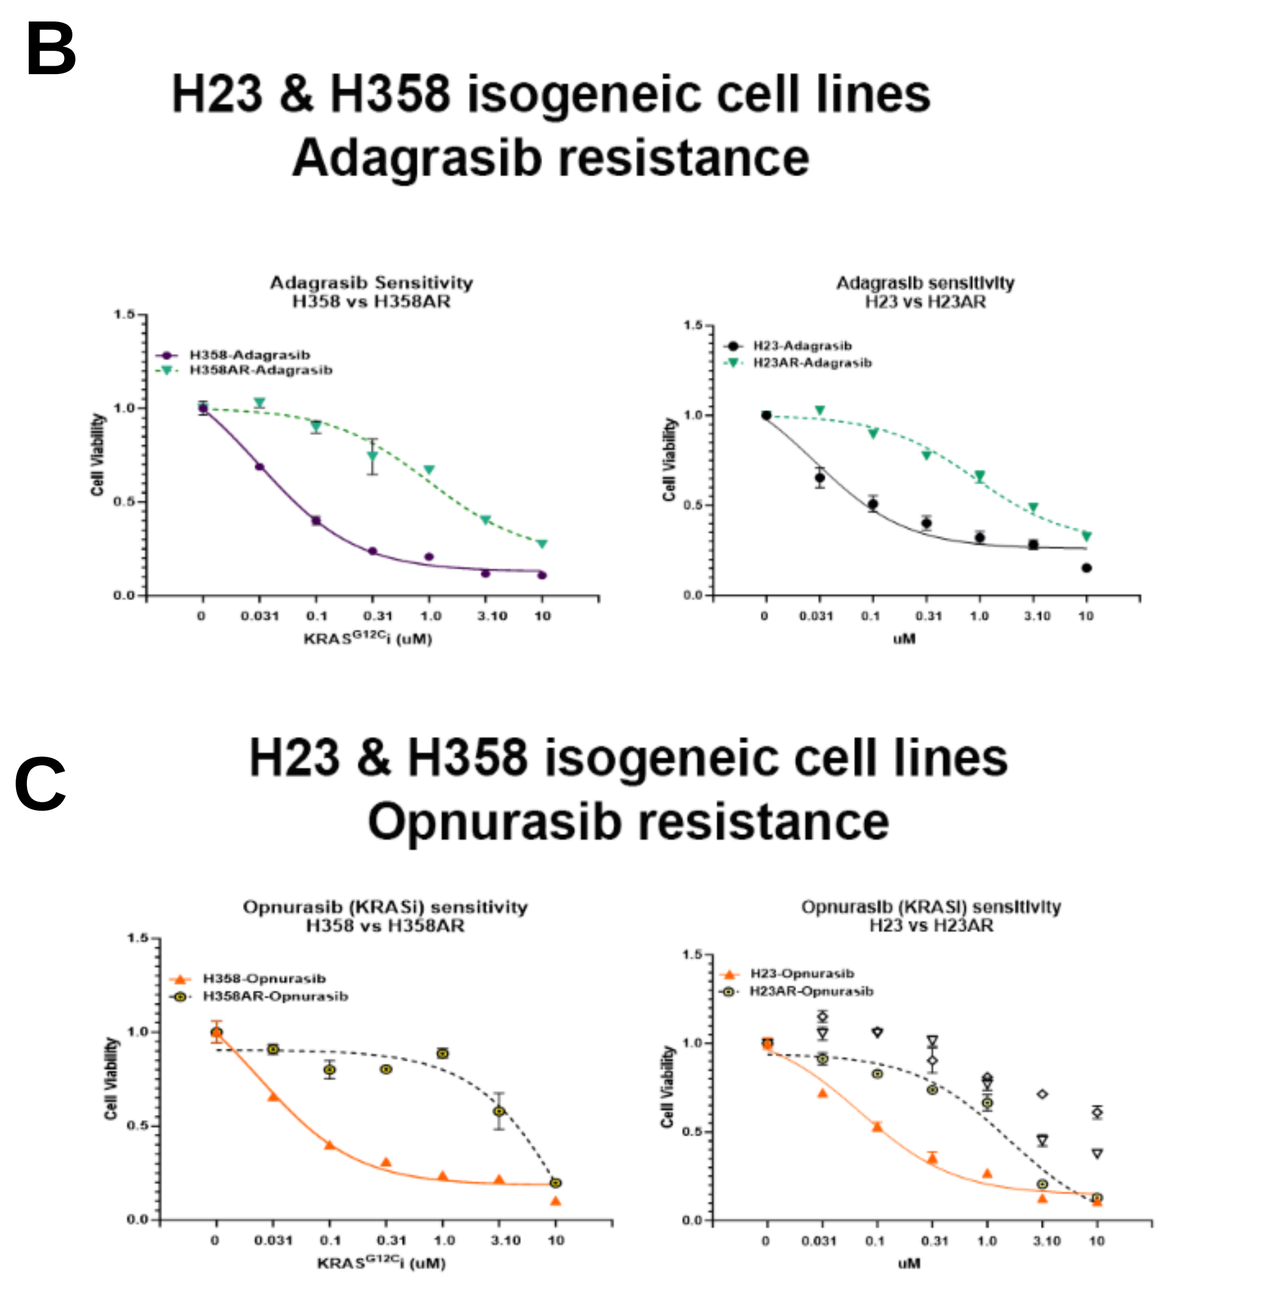
B
H23 & H358 isogeneic cell lines
Adagrasib resistance
C
H23 & H358 isogeneic cell lines
Opnurasib resistance

Fig 2. Development of sotorasib acquired resistant isogenic cell lines. A) H23AR and H358AR cell lines with acquired resistance to sotorasib were developed which showed >200 fold resistance to their sensitive counterparts; B) sensitivity tested to adagrasib; C) Sensitivity to opnurasib

Development of sotorasib-resistant CDX and xenograft models

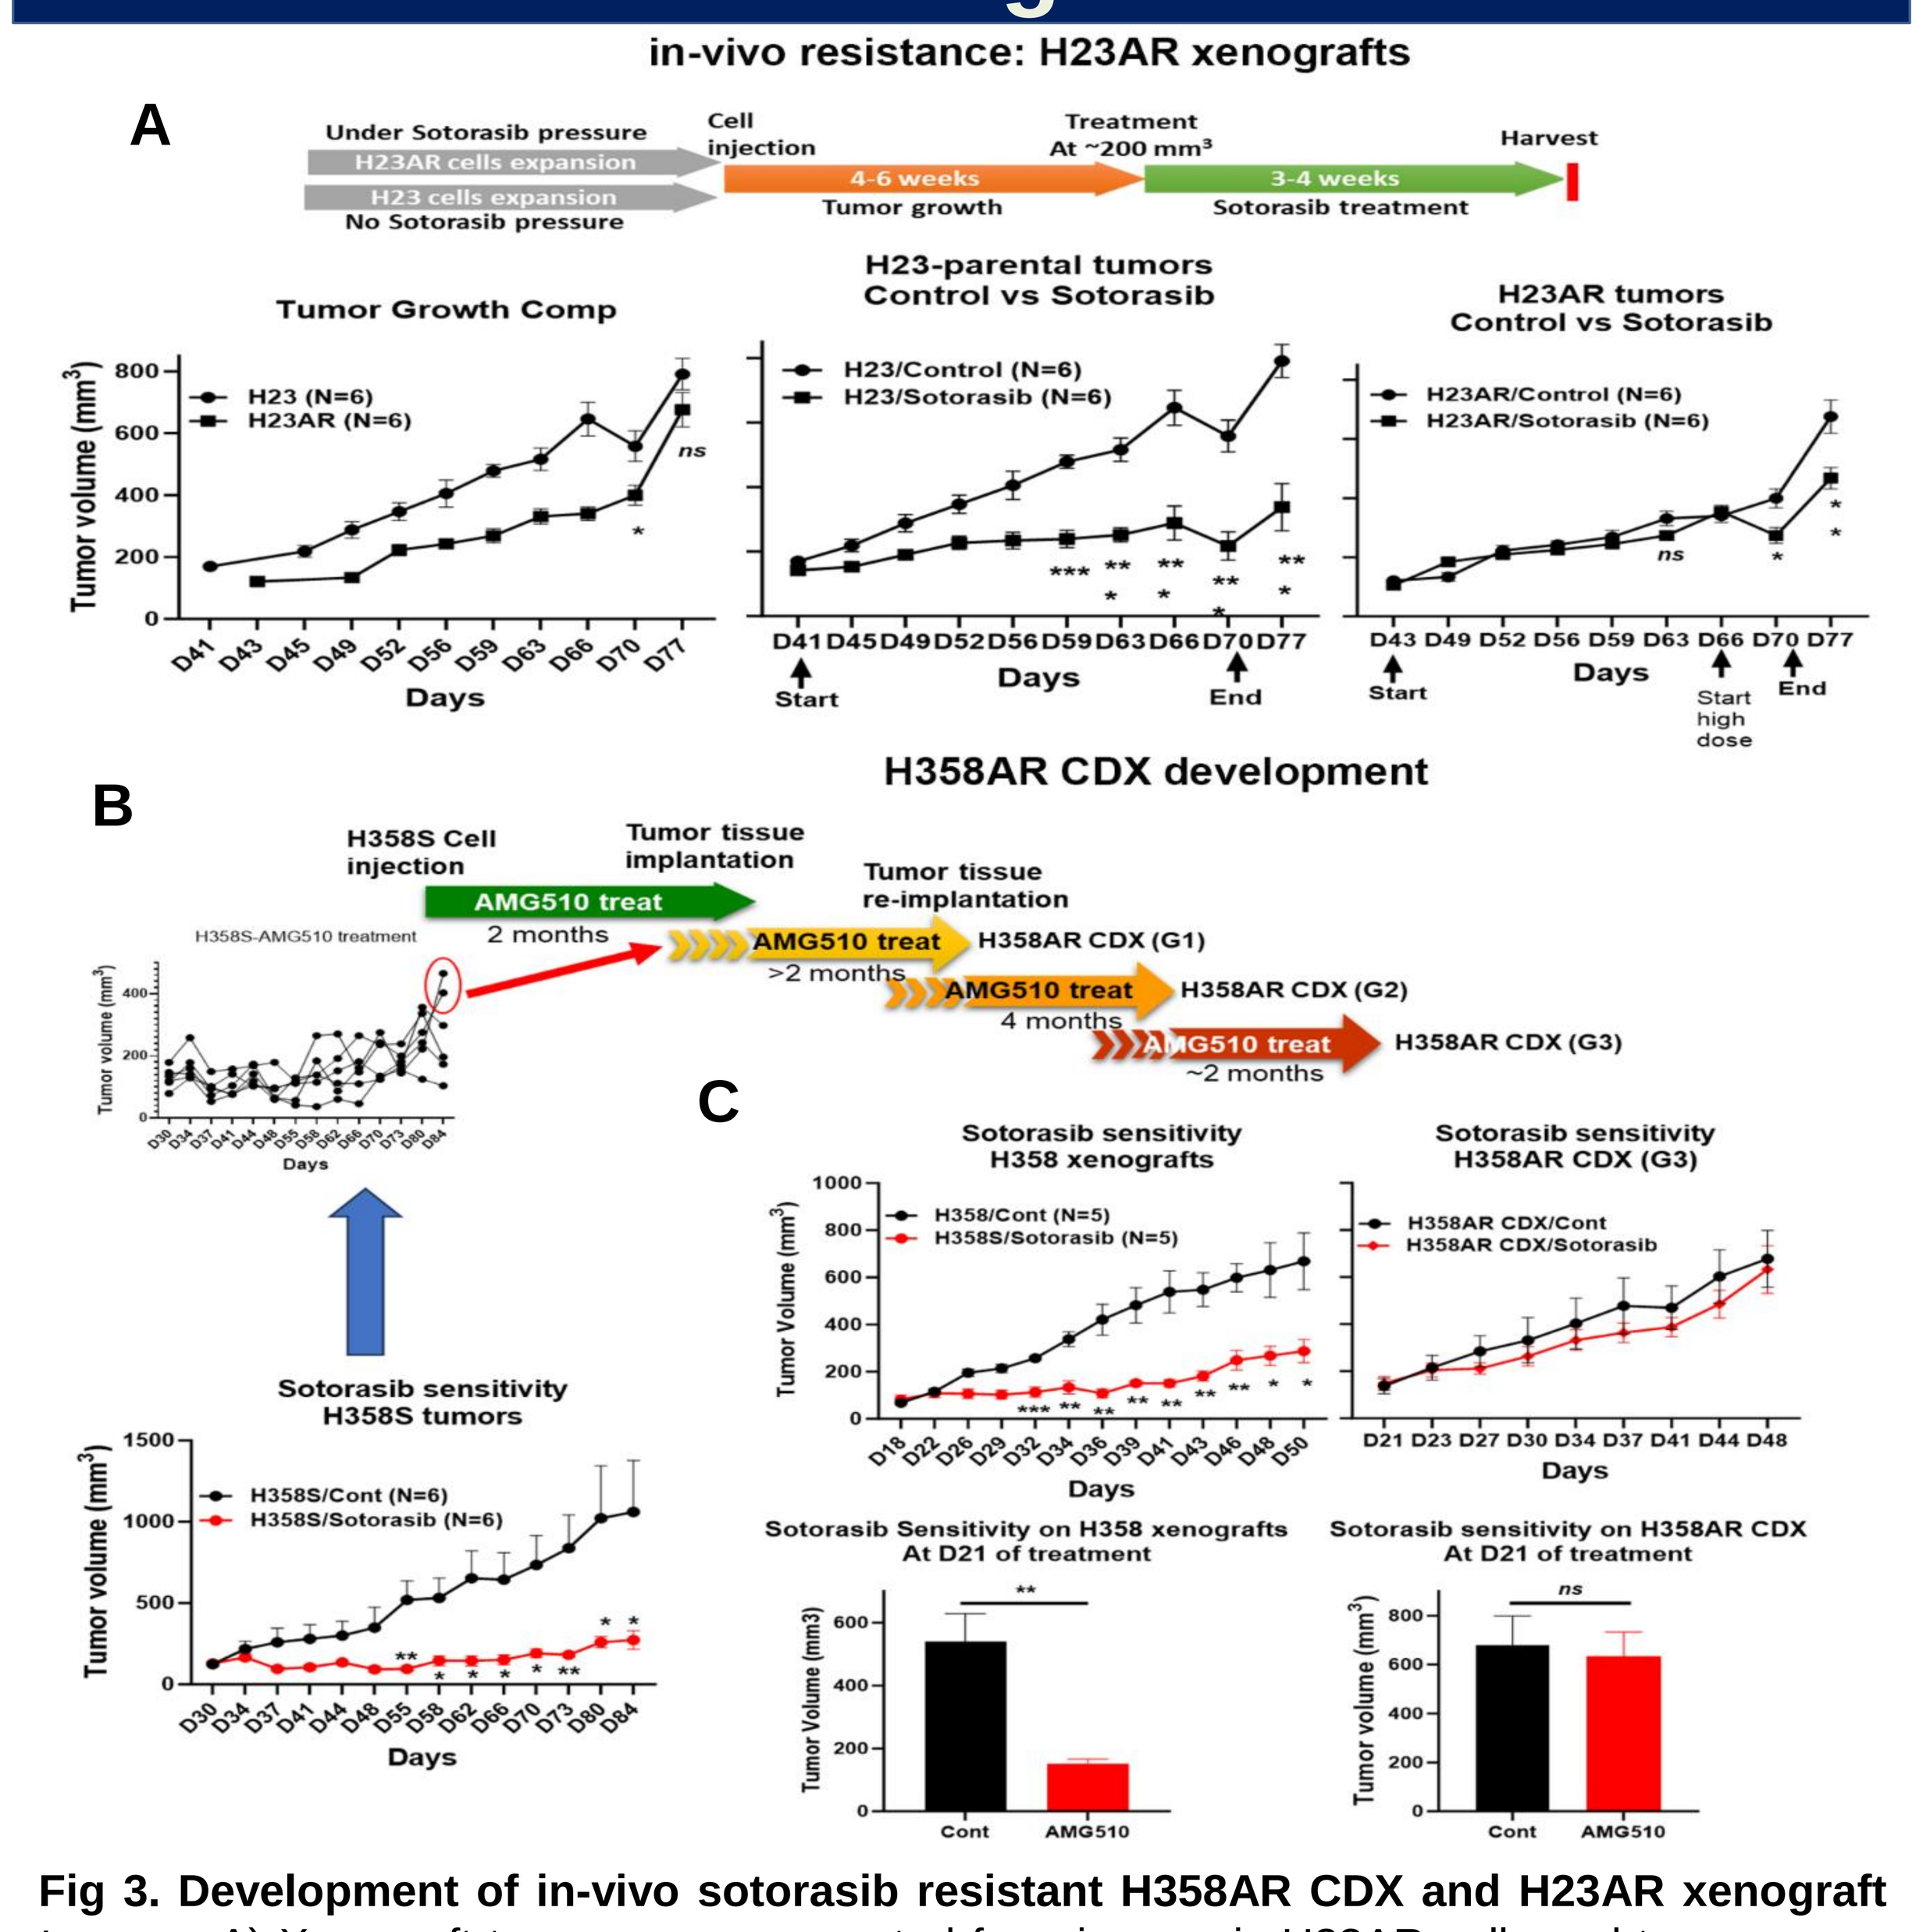
in-vivo resistance: H23AR xenografts
A
Under Sotorasib pressure
H23AR cells expansion
H23 cells expansion
No Sotorasib pressure
Cell injection
4-6 weeks
Tumor growth
Treatment At ~200 mm³
3-4 weeks
Sotorasib treatment
Harvest
Tumor Growth Comp
H23-parental tumors Control vs Sotorasib
H23AR tumors Control vs Sotorasib
H358AR CDX development
B
H358S Cell injection
Tumor tissue implantation
AMG510 treat
2 months
Tumor tissue re-implantation
H358AR CDX (G1)
H358AR CDX (G2)
H358AR CDX (G3)
Sotorasib sensitivity H358S tumors
C
Sotorasib sensitivity H358 xenografts
Sotorasib sensitivity H358AR CDX (G3)
Sotorasib Sensitivity on H358 xenografts At D21 of treatment
Sotorasib sensitivity on H358AR CDX At D21 of treatment

Fig 3. Development of in-vivo sotorasib resistant H358AR CDX and H23AR xenograft tumors. A) Xenograft tumors were generated from isogeneic H23AR cells and tumors were treated with sotorasib and the antitumor effect was compared; B) H358AR CDX was developed from H358 sensitive tumors by prolonged treatment with sotorasib for three passages to generate H358AR CDX (G3); C) G3 H358AR CDXs were reimplanted and treated with sotorasib to confirm sotorasib resistance in H358AR CDXs. * means p < 0.05, ** means p<0.005, and *** means p < 0.0005

Reactivation of KRAS signaling in acquired resistant PDXs

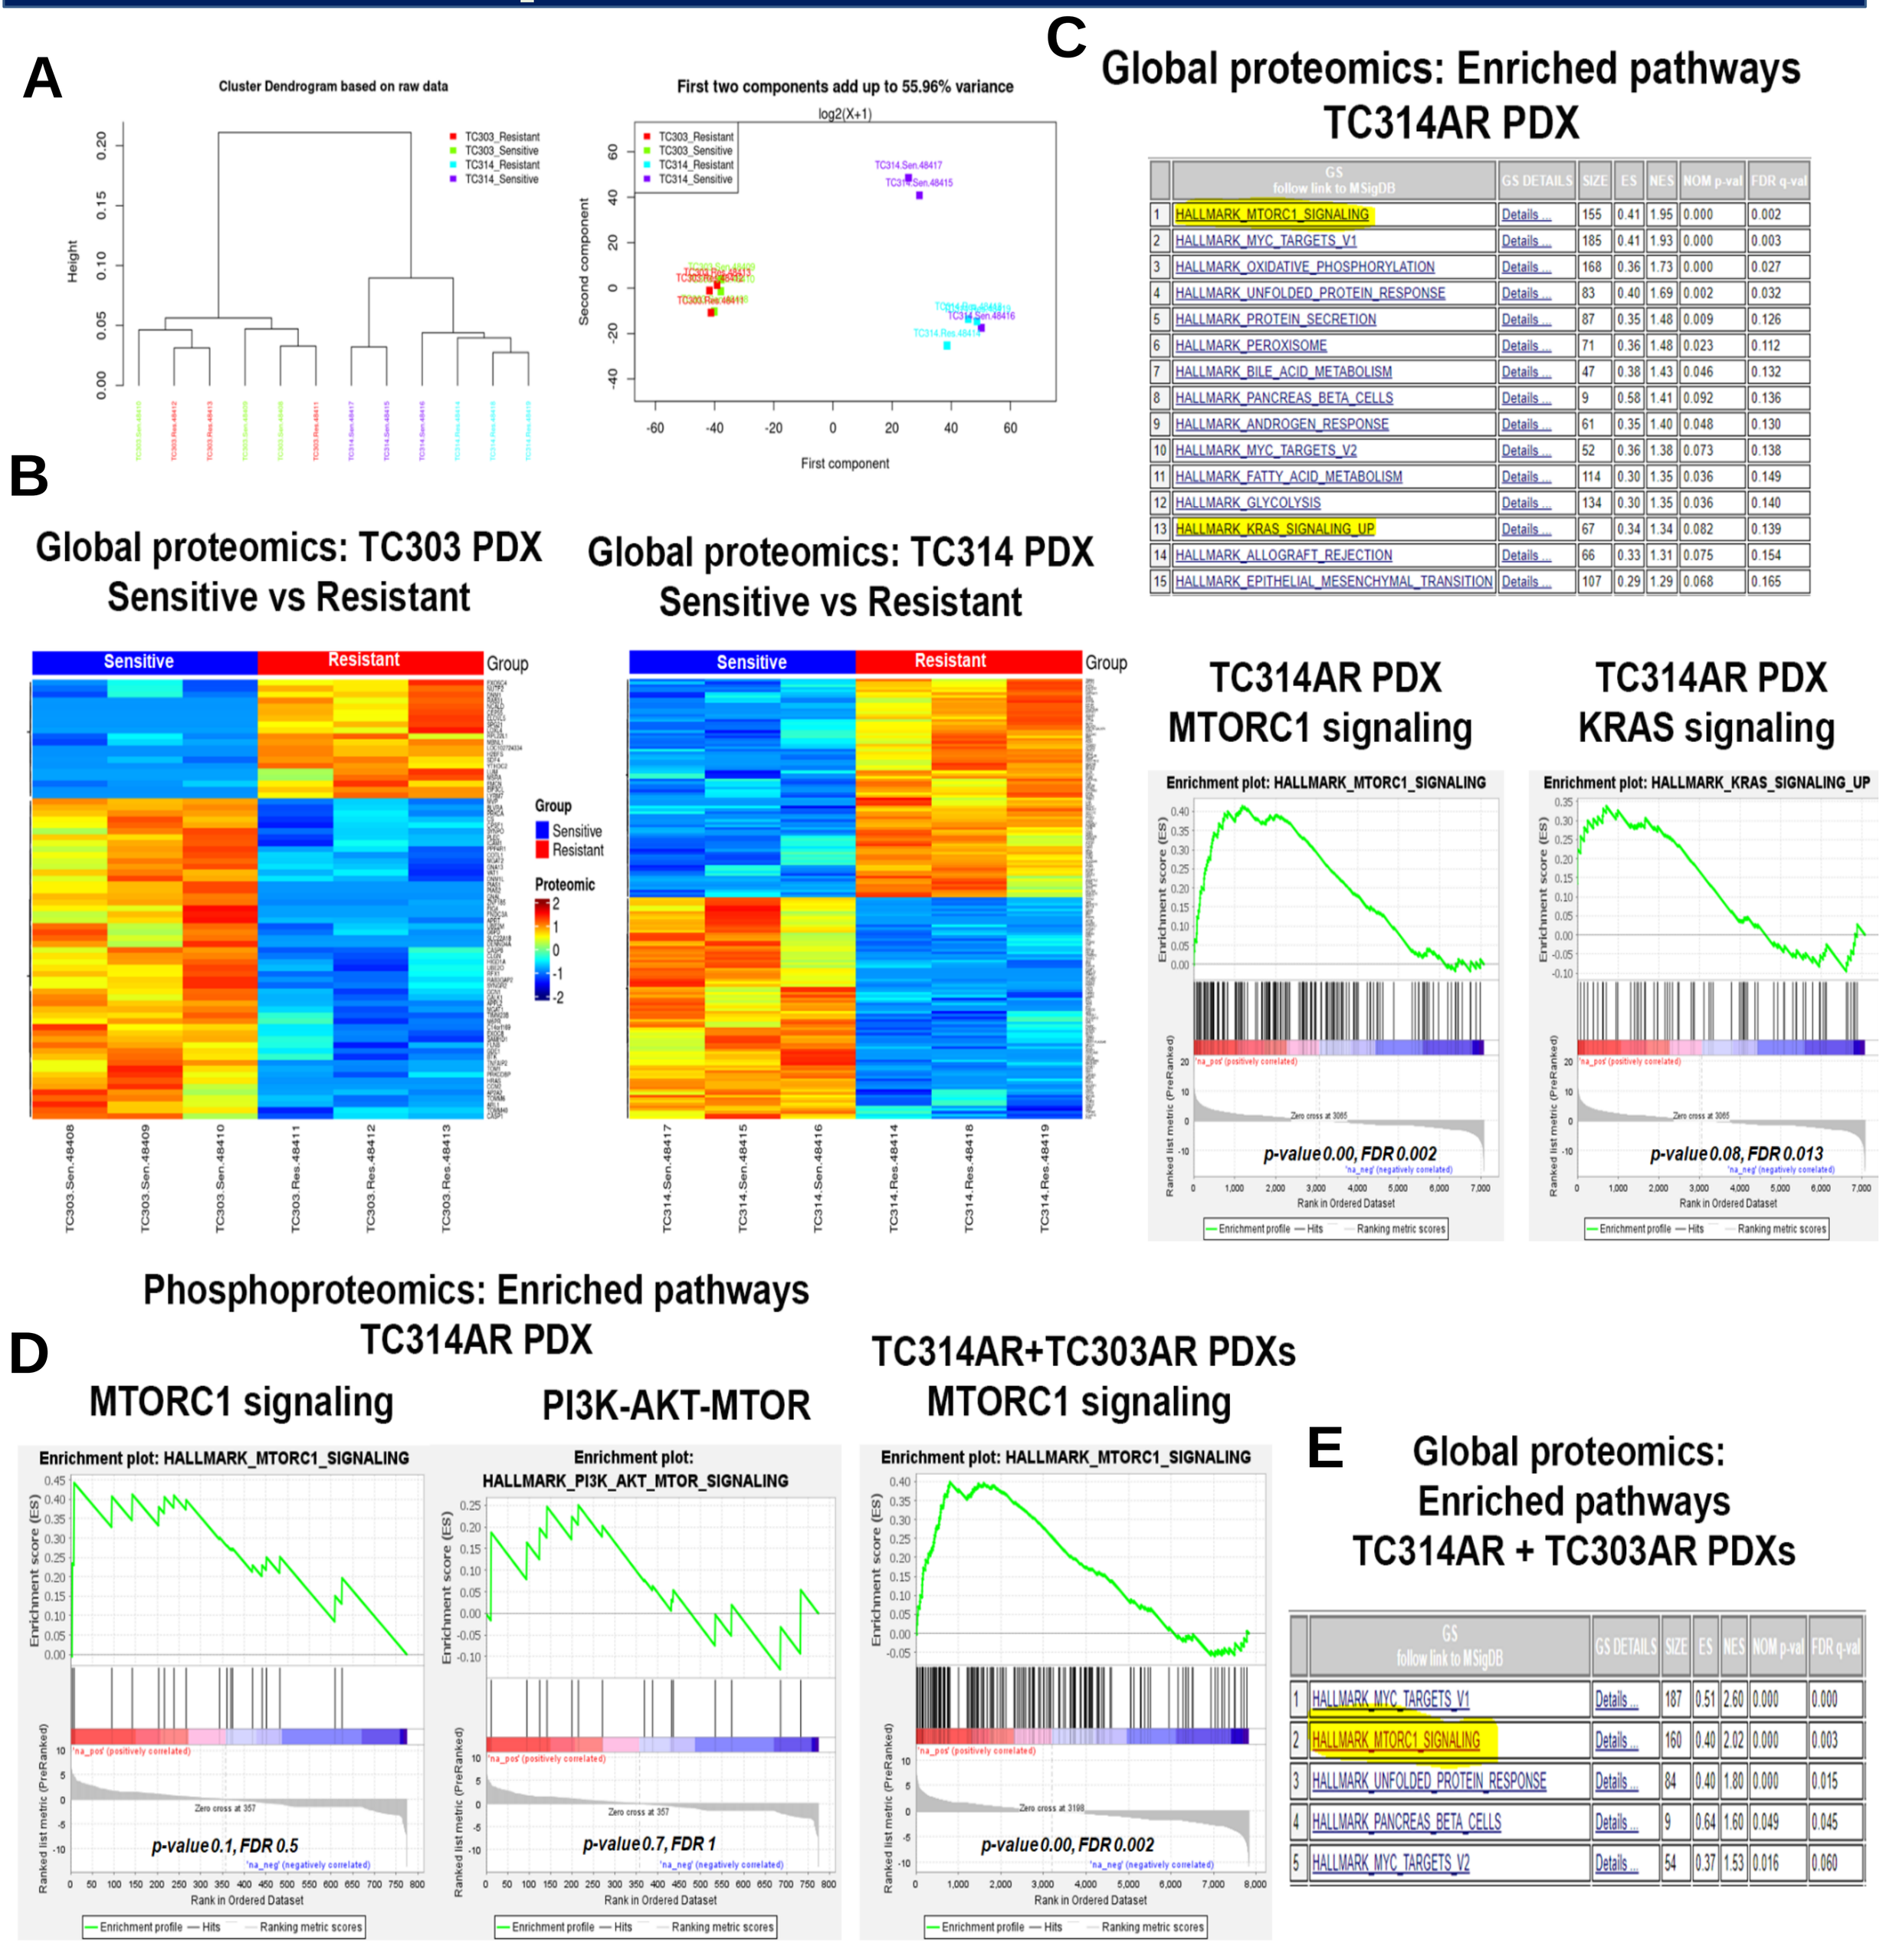
A
Cluster Dendrogram based on raw data
First two components add up to 55.96% variance
B
Global proteomics: TC303 PDX Sensitive vs Resistant
Global proteomics: TC314 PDX Sensitive vs Resistant
C
Global proteomics: Enriched pathways TC314AR PDX
TC314AR PDX MTORC1 signaling
TC314AR PDX KRAS signaling
p-value 0.00, FDR 0.002
p-value 0.08, FDR 0.013
D
Phosphoproteomics: Enriched pathways TC314AR PDX
MTORC1 signaling
PI3K-AKT-MTOR
TC314AR+TC303AR PDXs MTORC1 signaling
p-value 0.1, FDR 0.5
p-value 0.7, FDR 1
p-value 0.00, FDR 0.002
E
Global proteomics: Enriched pathways TC314AR + TC303AR PDXs

Fig 4. Global proteomics and phosphoproteomics on acquired resistant PDXs. A) A dendrogram and component curves showed the differences among PDX samples; B) Heatmaps showed the upregulation of a distinct set of proteins in AR PDXs (TC303AR & TC314AR) as compared with their sensitive PDXs; C-D) Enrichment analysis in global and phosphoproteomics in TC314AR showed MTORC1 & KRAS and MTORC1 & PI3K-Akt-MTOR pathways significantly upregulated respectively; E) MTORC1 signaling was also upregulated in TC303AR+TC314AR combined proteomics data;

TUSC2 inhibits colony formation & induces apoptosis in acquired resistant NSCLC cells

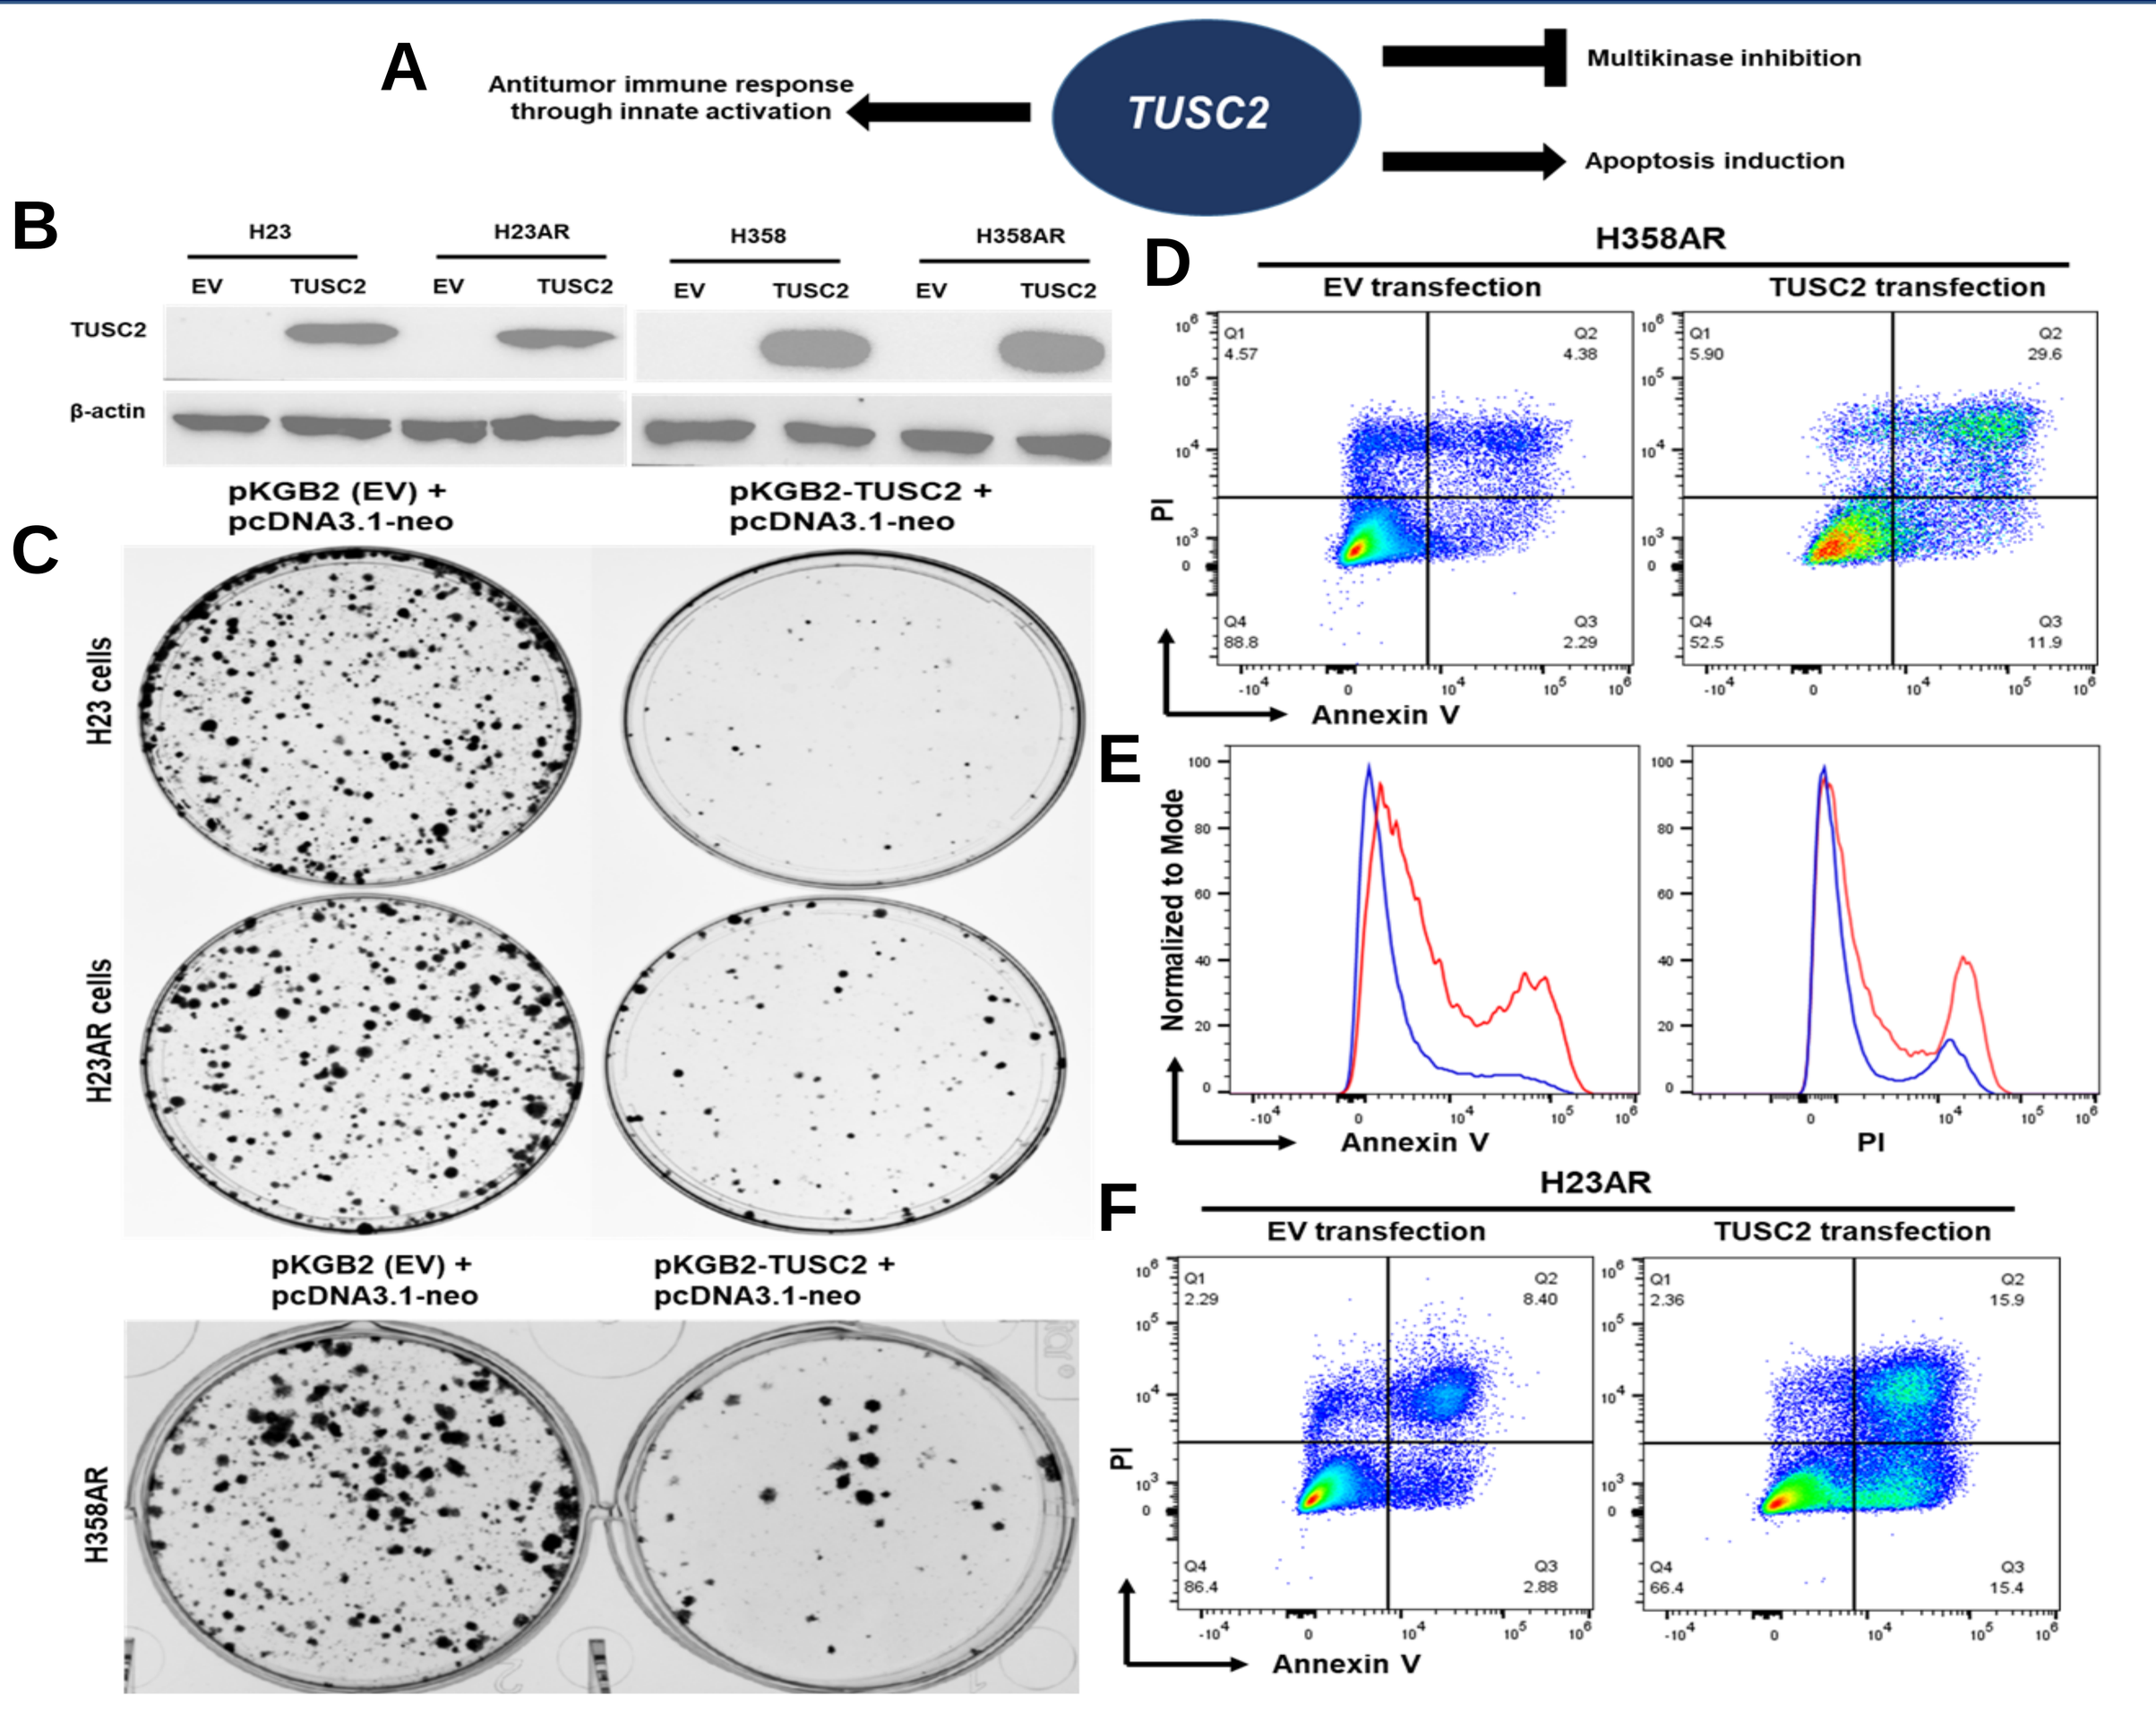
A
Antitumor immune response through innate activation
TUSC2
Multikinase inhibition
Apoptosis induction
B
H23
H23AR
H358
H358AR
EV
TUSC2
β-actin
C
pKGB2 (EV) + pcDNA3.1-neo
pKGB2-TUSC2 + pcDNA3.1-neo
H23 cells
H23AR cells
H358AR
D
H358AR
EV transfection
TUSC2 transfection
Annexin V
E
Normalized to Mode
Annexin V
PI
F
H23AR
EV transfection
TUSC2 transfection
Annexin V

Fig 5. TUSC2 inhibited colony formation and induced apoptosis in resistant cells. A) Mode of action of TUSC2; B) Transient transfection of TUSC2 in AR cells; C) Colony formation assay on stably TUSC2 transfected AR cells; D-F) Apoptosis of acquired resistant H23AR and H358AR cells after TUSC2 transfection.

Antitumor effect of TUSC2 on acquired resistant H23AR xenograft and TC314AR PDX models

H23AR xenograft model

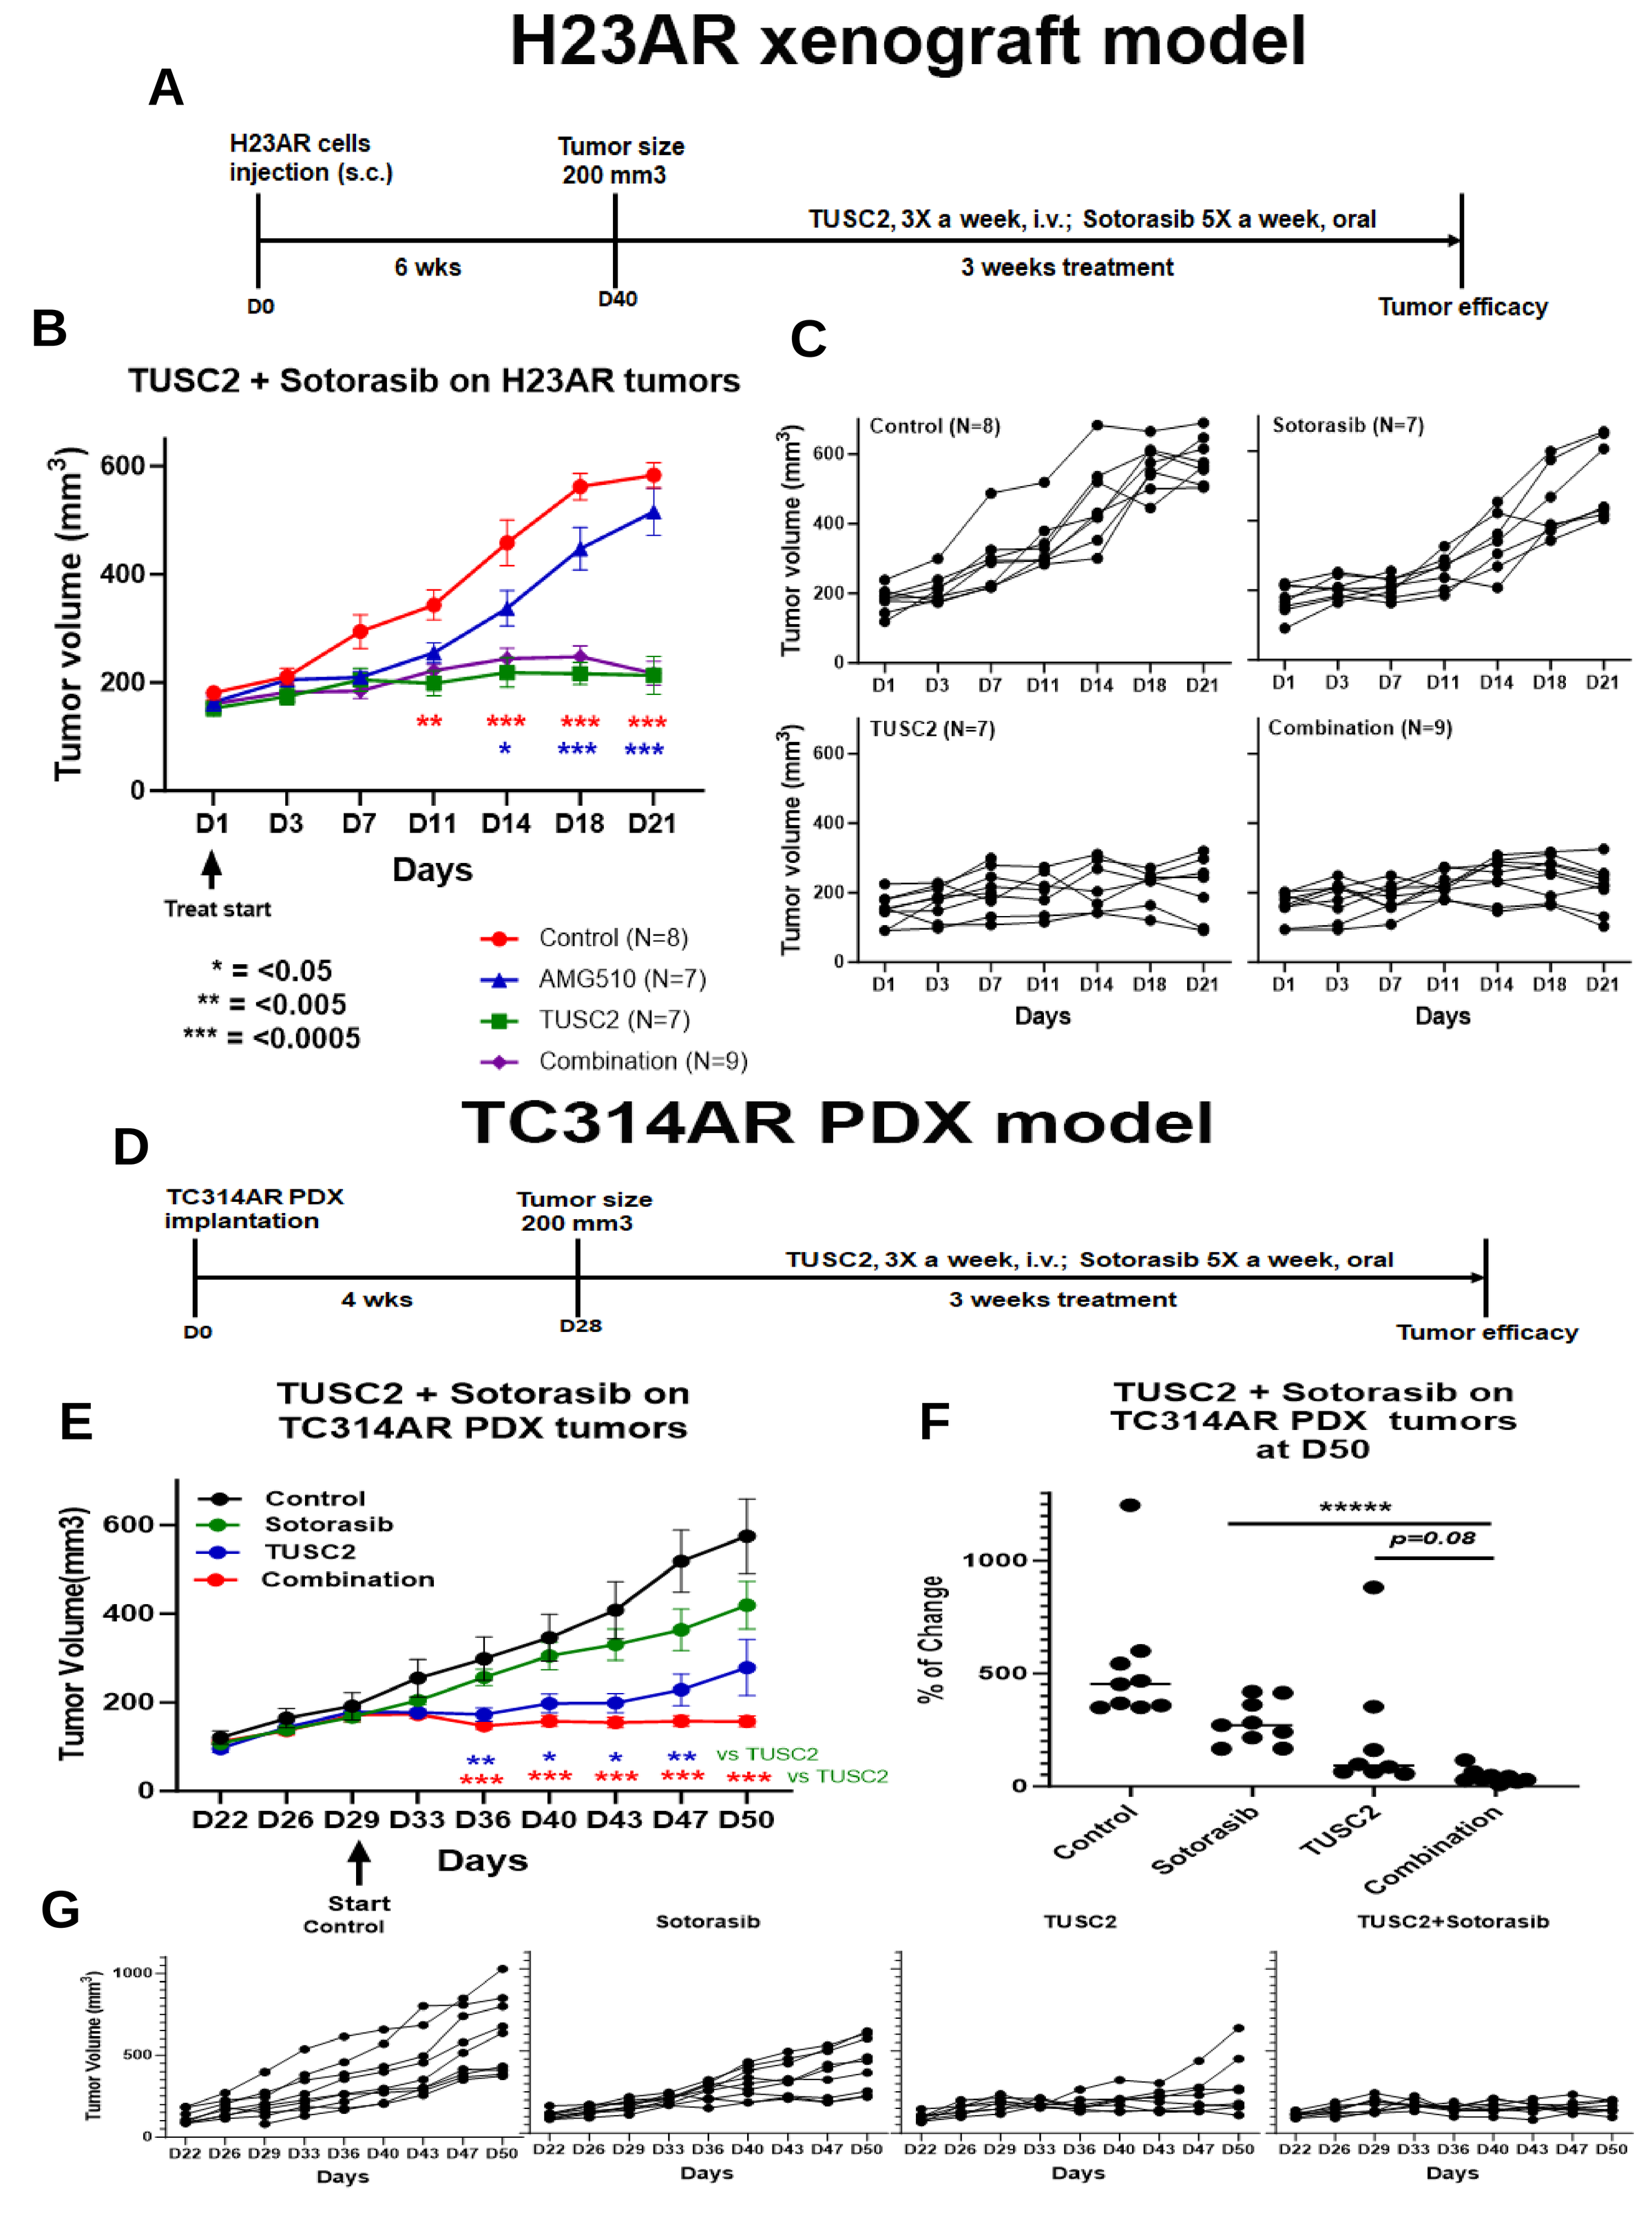
A
H23AR cells injection (s.c.)
Tumor size 200 mm3
TUSC2, 3X a week, i.v.; Sotorasib 5X a week, oral
6 wks
3 weeks treatment
Tumor efficacy
D0
D40
B
TUSC2 + Sotorasib on H23AR tumors
Treat start
* = <0.05
** = <0.005
*** = <0.0005
Control (N=8)
AMG510 (N=7)
TUSC2 (N=7)
Combination (N=9)
C
Control (N=8)
Sotorasib (N=7)
TUSC2 (N=7)
Combination (N=9)
TC314AR PDX model
D
TC314AR PDX implantation
Tumor size 200 mm3
TUSC2, 3X a week, i.v.; Sotorasib 5X a week, oral
4 wks
3 weeks treatment
Tumor efficacy
D0
D28
E
TUSC2 + Sotorasib on TC314AR PDX tumors
Control
Sotorasib
TUSC2
Combination
vs TUSC2
Start Control
F
TUSC2 + Sotorasib on TC314AR PDX tumors at D50
p=0.08
G
Control
Sotorasib
TUSC2
TUSC2+Sotorasib

Fig 6. Antitumor effect of TUSC2 gene therapy on H23AR xenografts and TC314AR PDXs. A) Treatment strategy for H23AR model; B) Combination effects on H23AR xenograft tumors; C) Individual mouse response to treatments; D) Treatment strategy for TC314AR PDXs; E) Antitumor effect of TUSC2, sotorasib and their combination; F) Percentage of changes in tumor volume after treatment; G) Individual mouse response to treatments. * means p < 0.05, ** means p<0.005, and *** means p < 0.0005

TUSC2 overcomes AR resistance by inducing antitumor immunity in a Humanized Mouse Model

TC314AR PDX on Humanized Mice

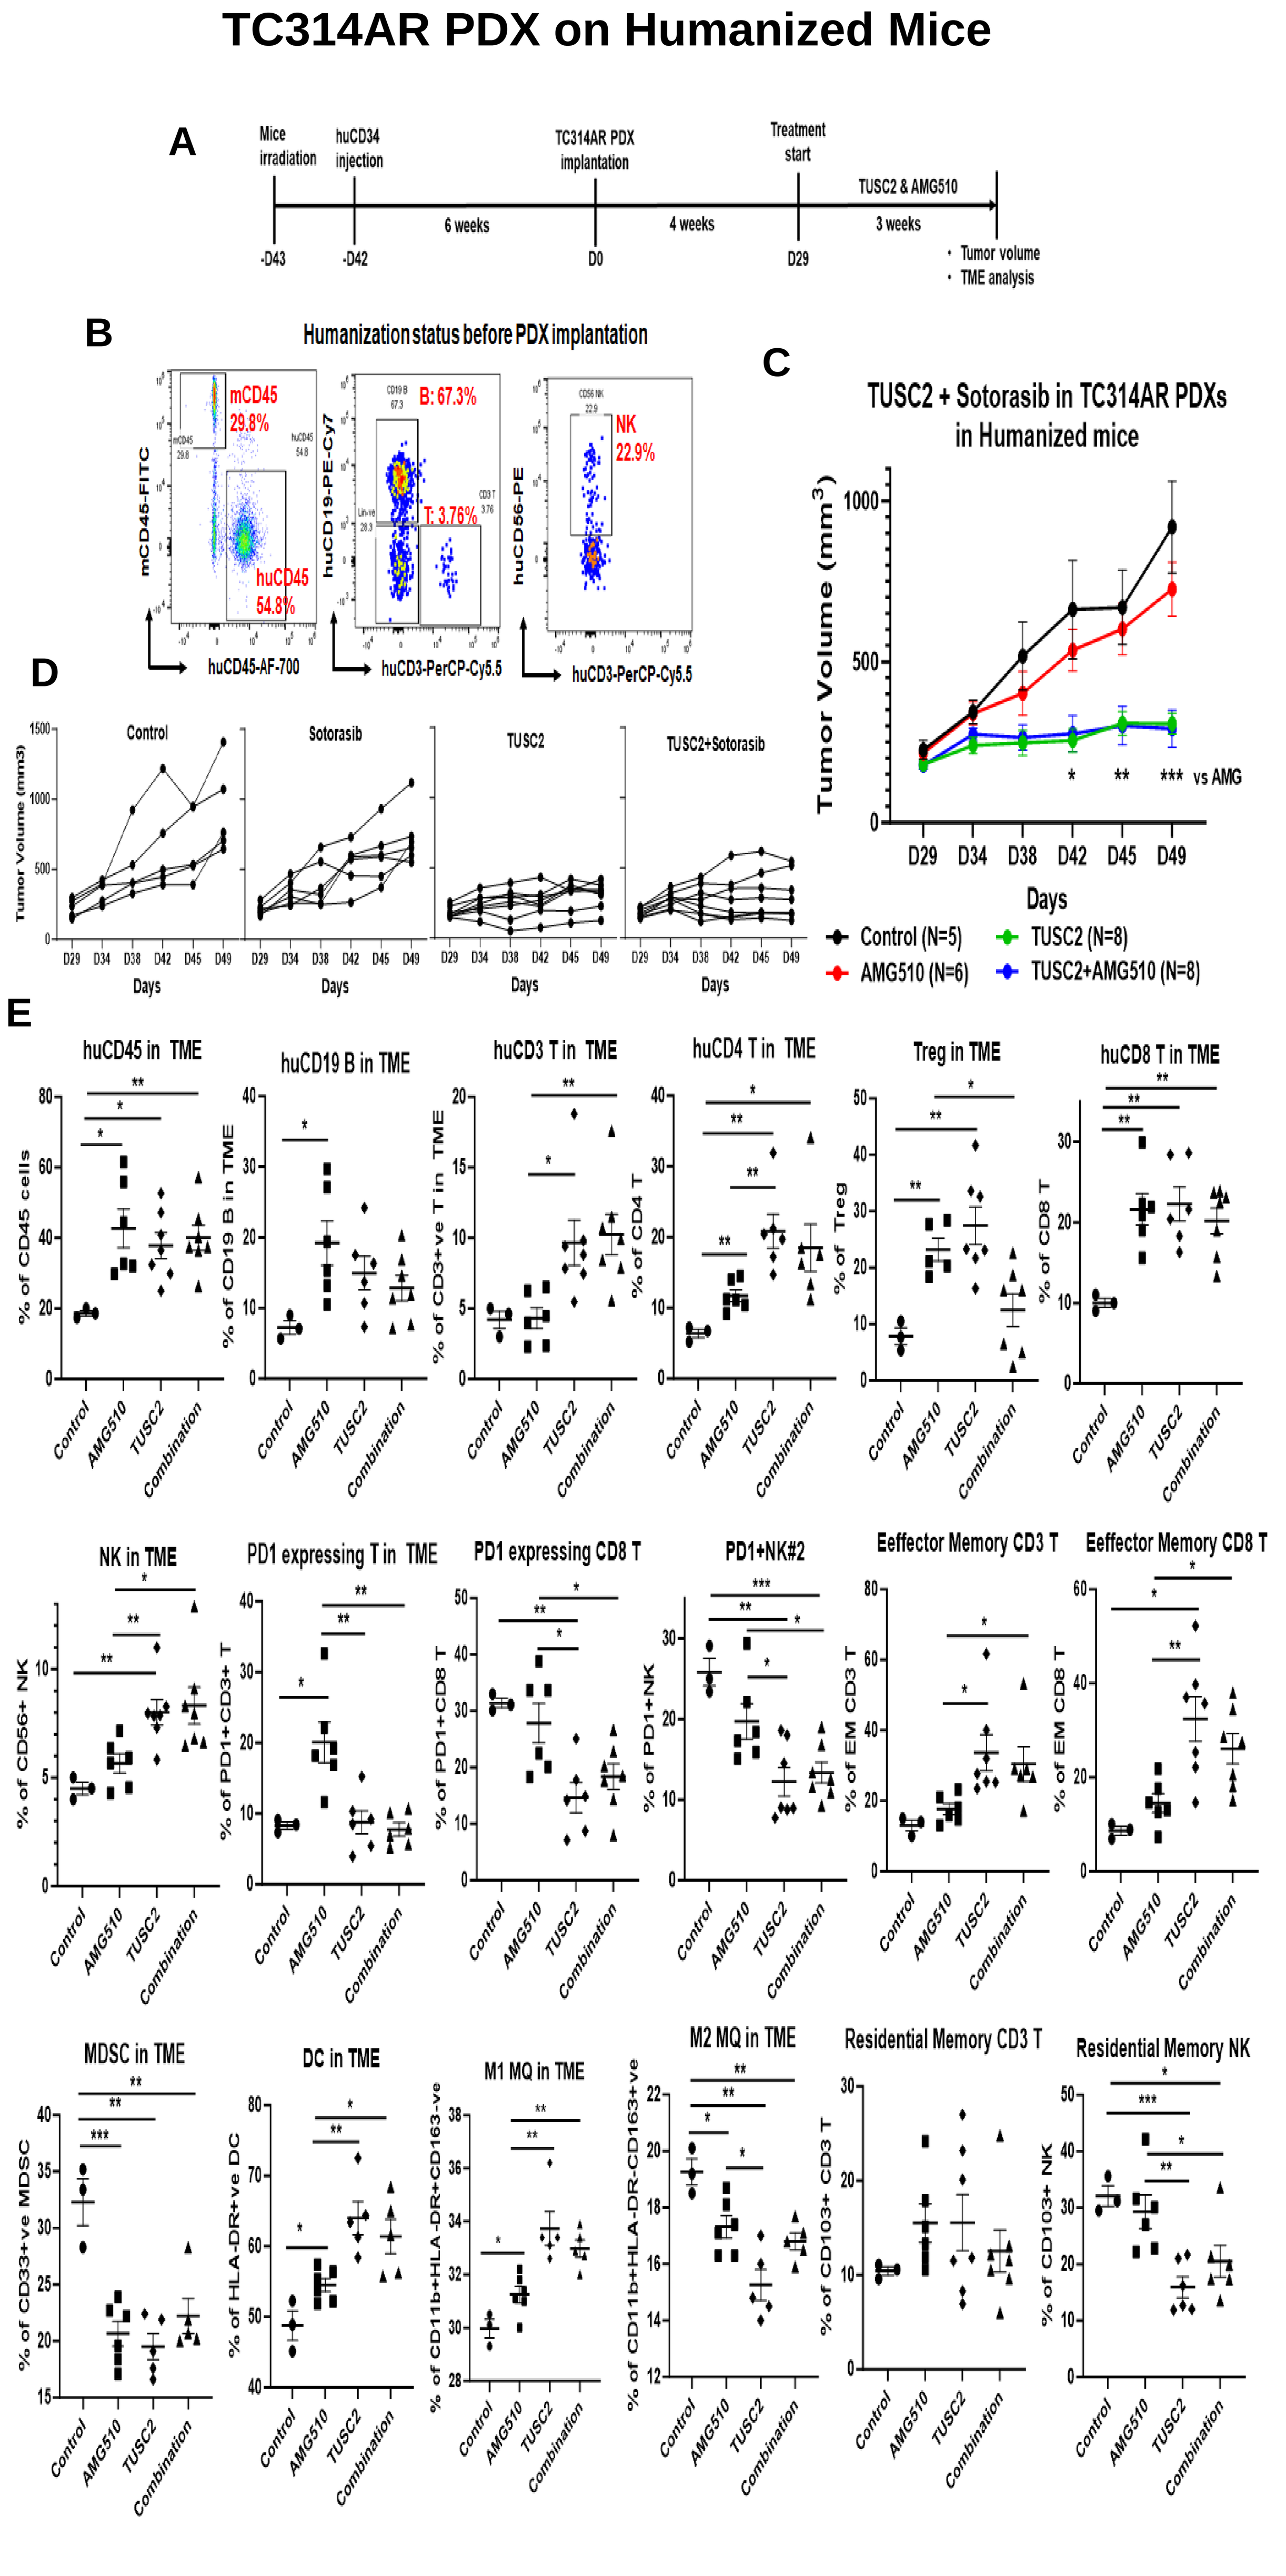
A
Mice irradiation
huCD34 injection
TC314AR PDX implantation
Treatment start
TUSC2 & AMG510
6 weeks
4 weeks
3 weeks
-D43
-D42
D0
D29
Tumor volume
TME analysis
B
Humanization status before PDX implantation
mCD45 29.8%
huCD45 54.8%
B: 67.3%
T: 3.76%
NK 22.9%
C
TUSC2 + Sotorasib in TC314AR PDXs in Humanized mice
vs AMG
Control (N=5)
AMG510 (N=6)
TUSC2 (N=8)
TUSC2+AMG510 (N=8)
D
Control
Sotorasib
TUSC2
TUSC2+Sotorasib
E
huCD45 in TME
huCD19 B in TME
huCD3 T in TME
huCD4 T in TME
Treg in TME
huCD8 T in TME
NK in TME
PD1 expressing T in TME
PD1 expressing CD8 T
PD1+NK#2
Eeffector Memory CD3 T
Eeffector Memory CD8 T
MDSC in TME
DC in TME
M1 MQ in TME
M2 MQ in TME
Residential Memory CD3 T
Residential Memory NK

Fig 7. The antitumor immune response of TUSC2 on TC314AR PDXs in humanized mice. A) experimental strategy; B) Humanization status before PDXs implantation; C) Antitumor effect of TUSC2, sotorasib (AMG510) and its combination; D) Individual mouse response towards treatment; E) Tumor microenvironment (TME) analysis in humanized mice: (upper panel) effect on human CD45, CD3 T, CD4 T, CD8 T, Treg; (middle panel) effect on human NK, PD1+CD3 T, PD1+CD8 T, PD1+NK, Effector memory CD3 & CD8 T cells; (bottom panel) effect on human MDSC, DC, M1 & M2 MQ, Residential memory CD3 T and residential memory NK cells. * means p < 0.05, ** means p<0.005, and *** means p < 0.0005

Conclusions

- Sotorasib-resistant patient-derived xenografts (PDX) and cell line-derived xenografts (CDX) were developed, showing no antitumor effect of sotorasib.
- Two isogeneic acquired resistant NSCLC cell lines were generated and both AR cell lines showed >100-fold resistance over sensitive counterparts
- No additional KRAS mutation was found; instead reactivation of the KRAS pathway and upregulation of PI3K-AKT-mTOR signaling were found to be the main mechanisms of acquired resistance.
- Restoration of TUSC2, a multipotent tumor suppressor, by transient transfection significantly reduced colonly formation and induced apoptosis in acquired resistant cells
- Robust antitumor activity was found on acquired resistant H23AR xenograft tumors when tumors were treated with quaratusugene ozeplasmid, a lipoplex gene therapy containing the TUSC2 gene
- TUSC2 gene therapy also showed a synergistic antitumor effect on acquired resistant TC314AR PDX tumors when PDXs are treated with TUSC2 and sotorasib combination
- quaratusugene ozeplasmid exhibited a strong antitumor effect on TC314AR PDXs which was significantly superior to sotorasib treatment in humanized mice
- TUSC2 generated a strong antitumor immune response in TC314AR PDXs in humanized mice which was associated with significantly increased infiltration of human CD3, CD4, cytotoxic T, NK cells and inhibition of human regulatory T cells
- PD1 expressing T and NK cells were significantly downregulated whereas effector memory CD3 and CD8 T cells were markedly increased by TUSC2 treatment.
- TUSC2 aactivated innate immunity by enhanced infiltration of DC and M1 macrophages with significant inhibition of MDSC and M2 macrophages.

Disclosures

Jack A. Roth is a consultant, stock owner (including pending patent) in Genprex, Inc. All other authors have declared that no competing interests exist.